#176001 Kungatsiajuk: Supporting the healthy smiles of NunatuKavut Children

This work was funded by CIHR-IAPH (246226) in partnership with the NunatuKavut Health and Social Sector

*A. DEMSEY[1], M. MCNALLY[1], D. MARTIN[2], D. WALL[3], **D. MATTHEWS**[1], H. CASTLEDEN[4], and I. WORDEN-DRISCOLL[2]*

1. Faculty of Dentistry, Dalhousie University, Halifax, NS Canada 2. School of Health and Human Performance, Dalhousie University, Halifax, NS Canada

3. NunatuKavut Social and Health Sector, Happy Valley - Goose Bay, NL Canada 4. Faculty of Management / Faculty of Medicine, Dalhousie University, Halifax, NS Canada

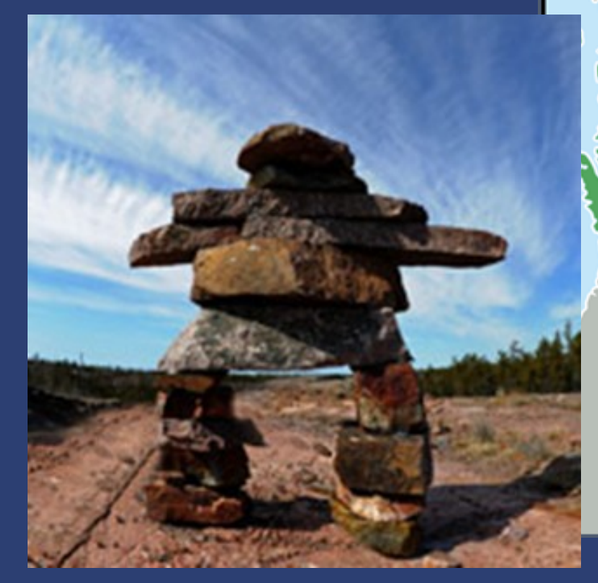

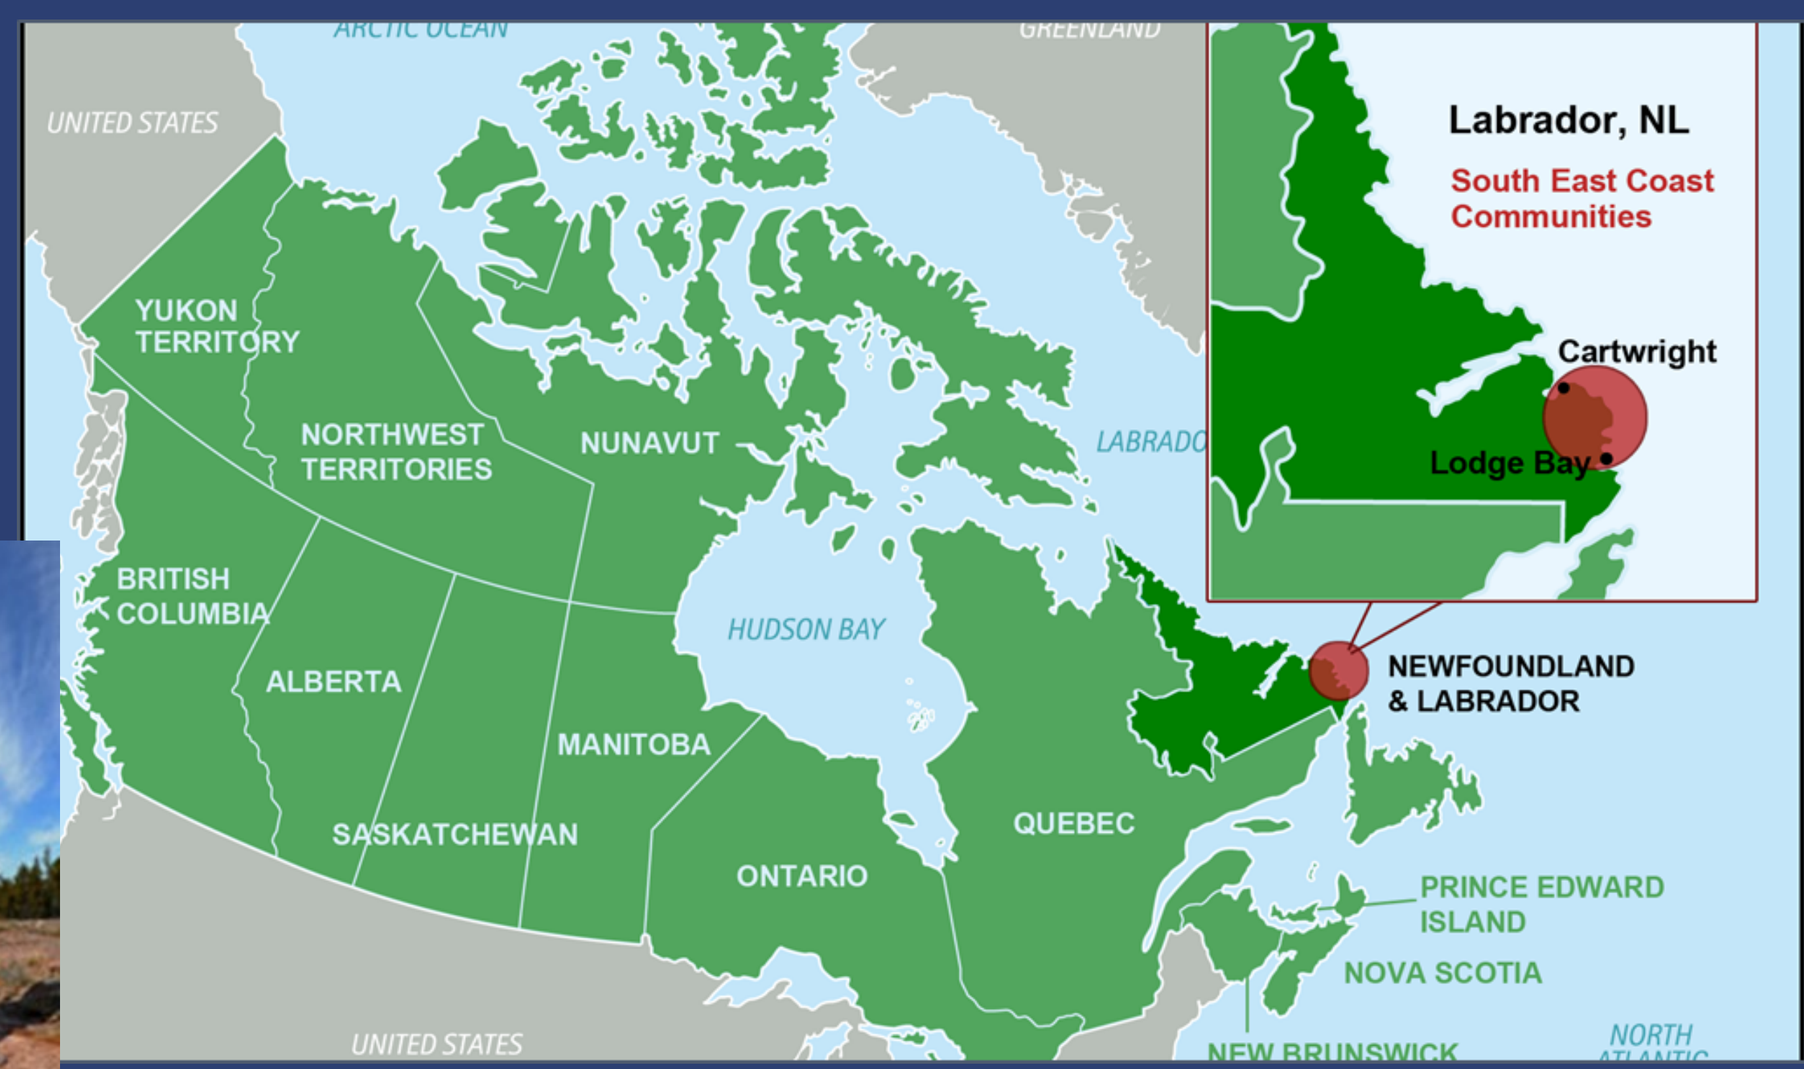
Labrador, NL
South East Coast Communities
Cartwright
Lodge Bay
YUKON TERRITORY
NORTHWEST TERRITORIES
NUNAVUT
BRITISH COLUMBIA
ALBERTA
MANITOBA
SASKATCHEWAN
ONTARIO
QUEBEC
HUDSON BAY
NEWFOUNDLAND & LABRADOR
PRINCE EDWARD ISLAND
NOVA SCOTIA
UNITED STATES

Purpose

To examine oral health status, perceptions regarding oral health and oral health care services for NunatuKavut children and adolescents residing in southeast Labrador, Canada.

Background

- An interdisciplinary community-based health planning workshop, *'Community Health Research in Labrador: Listening, Learning and Working Together'* (2006) identified poor oral health as among the top health priorities affecting NunatuKavut people in southeast Labrador, Canada. This finding provides the impetus for this research which focuses on the oral health of children and adolescents.
- Poor oral health negatively affects children's self-esteem, eating and sleeping habits and overall well-being.
- Compared to non-Aboriginal children, Canadian Aboriginal children are reported to suffer from significant oral health disparities.
- Inuit communities of NunatuKavut are geographically isolated, contributing to financial and logistical barriers for accessing health care.
- The Inuit of NunatuKavut do not receive health benefits offered by Health Canada's First Nations and Inuit Health.

Methods

The overall research design follows a community based participatory approach using mixed methodology. This report details the survey portion only.

Survey instrument:

A two part validated survey based on the Inuit Oral Health Survey (IOHS, 2007-2008) and Canadian Oral Health Measures Survey (CHMS, 2007-2009) was conducted in May 2012:

1. Structured interview of care-givers and/or children given by a trained community-based interviewer
 - Perceptions of oral health
 - Oral health care habits
 - Utilization of oral health services
 - Third party dental coverage
2. Clinical oral exam completed by one WHO calibrated dentist
 - Baseline oral health status
 - Treatment needs

Participants:

All children up to the age of 17 from 7 NunatuKavut communities in remote Labrador, Canada were invited to participate (n=378).

Due to geographic isolation and weather, 2 communities (n=41) were not accessible.

Results

Response Rate:
Clinical examination: 70.1% (n=265)
Structured survey: 66.4% (n=251)

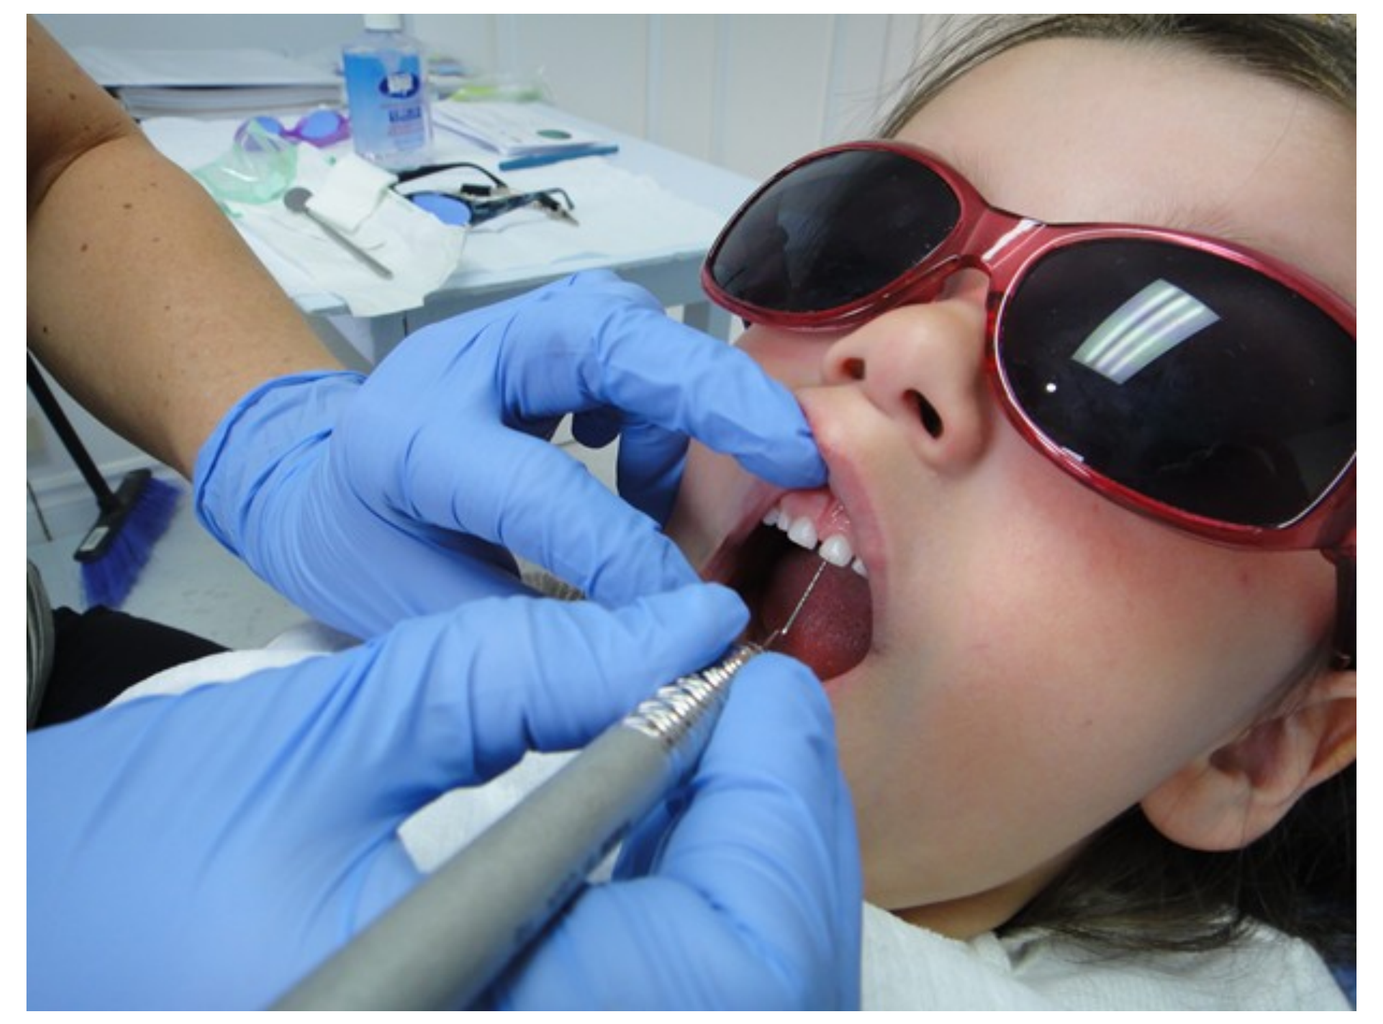

Table 1: Oral health related quality of life

	Age Group (%)		
Characteristic	**0 – 5 years**	**6 – 11 years**	**12 – 17 years**
Self / proxy-reported fair or poor oral health	7.4	8.4	7.8
Avoided foods because of problems with their mouth	14.8	10.5	8.8
Persistent pain or ongoing pain anywhere in their mouth	13.0	8.4	9.8
Time lost from normal activities, work or school activities in the past 12 months	24.1	31.6	37.3

Table 2: Preventive practices

	Age Group (%)		
Characteristic	**0 – 5 years**	**6 – 11 years**	**12 – 17 years**
Brushing 2 or more times per day	68.5	93.7	96.1
Flossing at least 5 times per week	7.4	16.8	26.5
Visits a dental professional at least once per year for check-ups	53.7	77.9	66.3

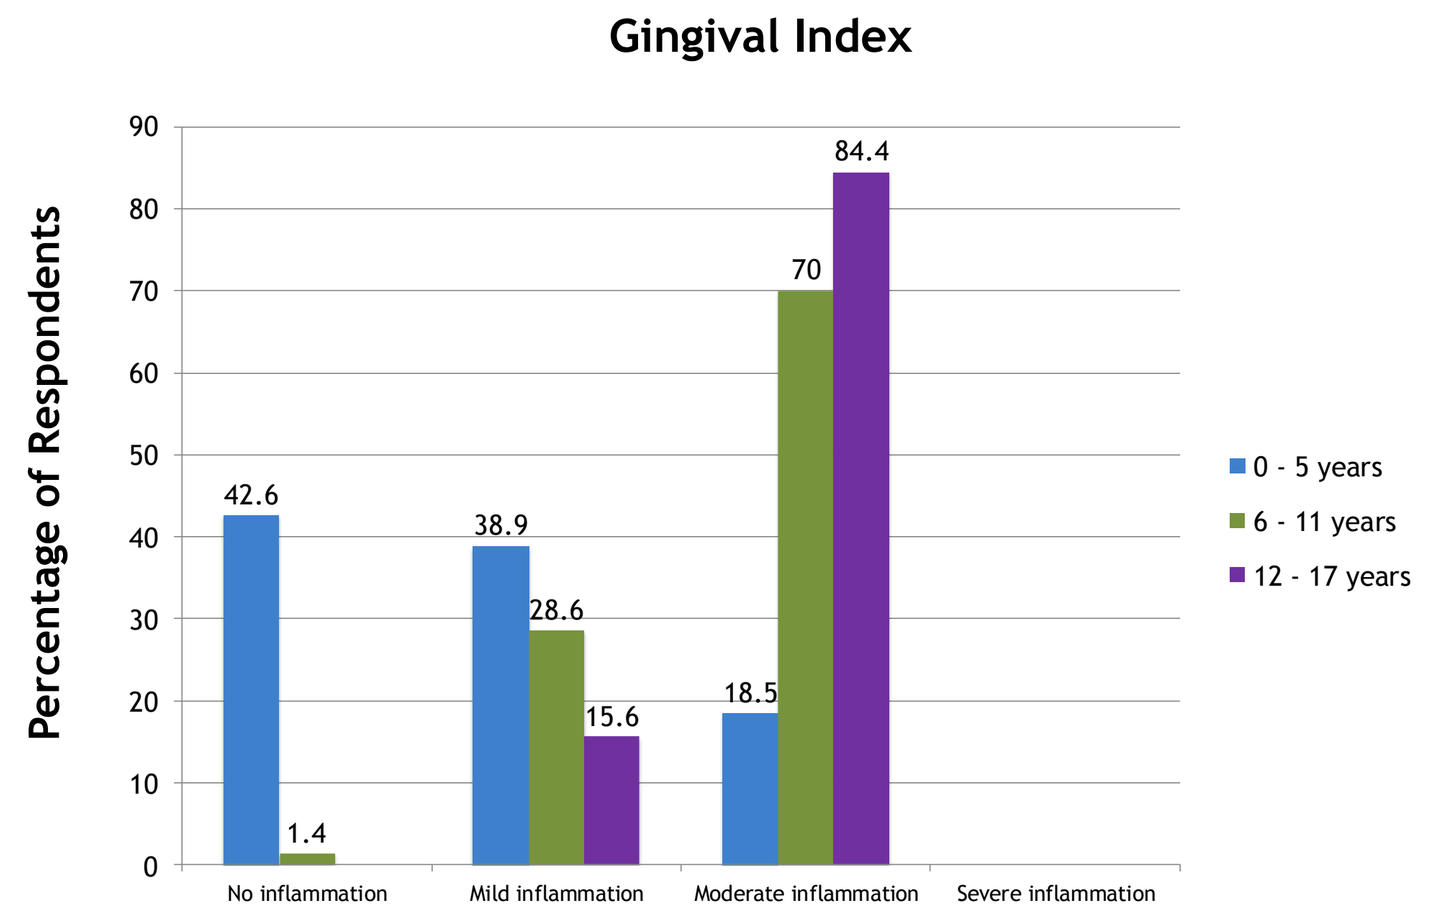
Gingival Index
Percentage of Respondents
0 - 5 years
6 - 11 years
12 - 17 years
42.6
1.4
38.9
28.6
15.6
18.5
70
84.4
No inflammation
Mild inflammation
Moderate inflammation
Severe inflammation

70.1% of respondents reported some form of third party dental insurance coverage. Of these:
- 49% (n= 87) were covered through a provincial plan
- 46% (n= 81) were covered through an employer sponsored plan
- 5% (n=10) were covered though social services (welfare)

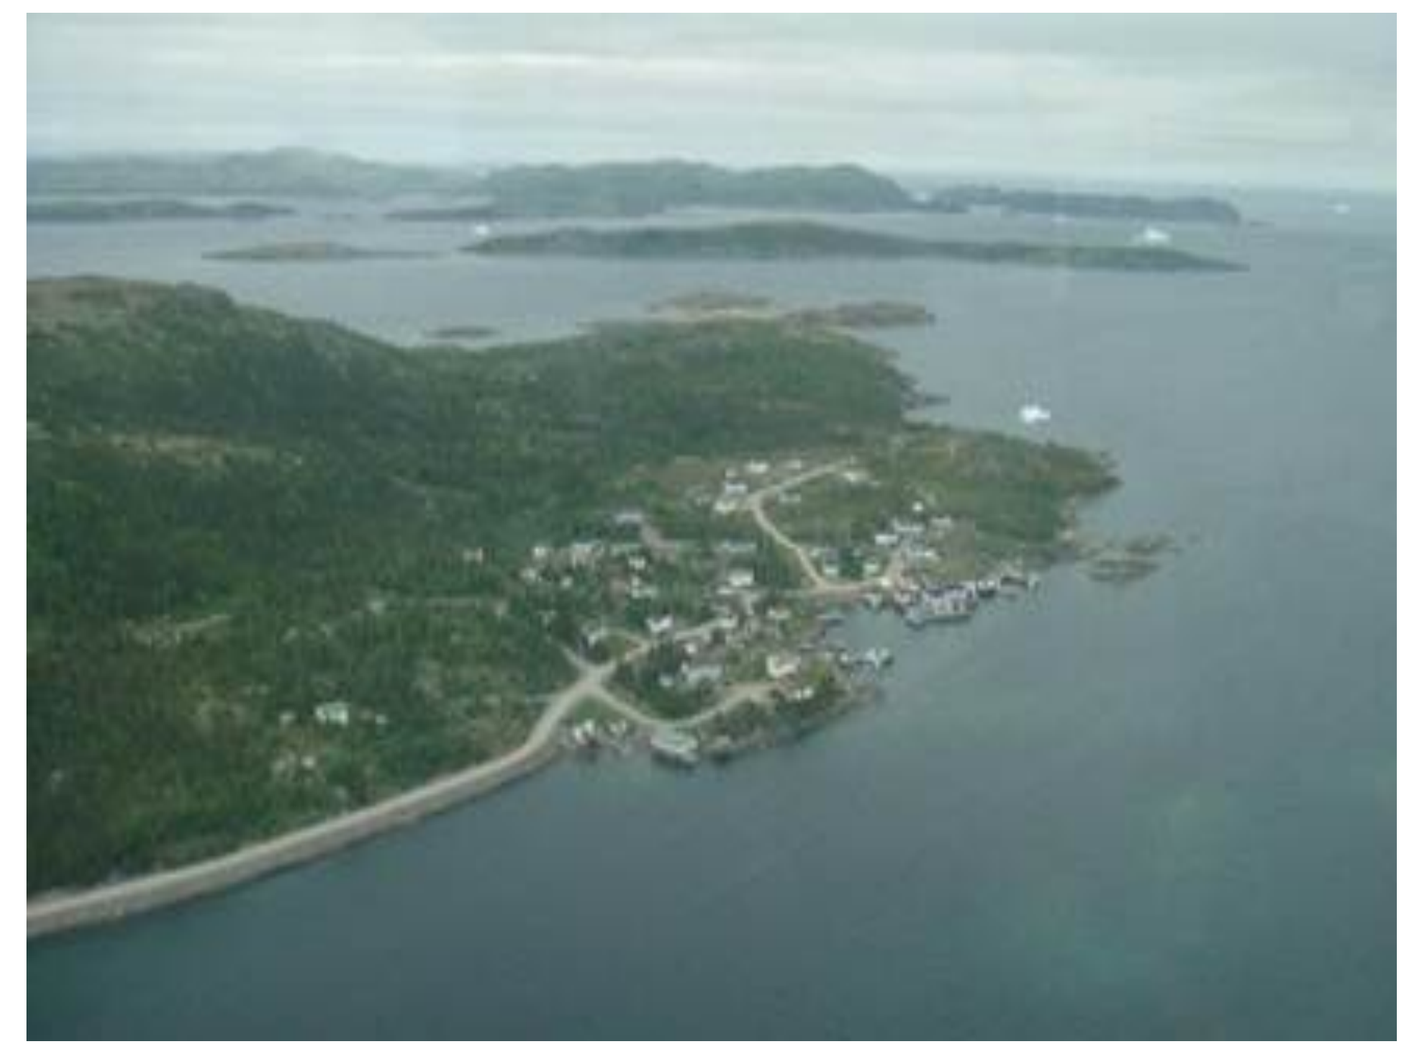

Table 3: Untreated coronal caries

	Age Group							
	0 – 5 years (Primary teeth)		**6 – 11 years (Primary teeth)**		**6 – 11 years (Primary & permanent teeth)**		**12 – 17 years (Permanent teeth)**	
Characteristic	% with 1 or more	Mean	% with 1 or more	Mean	% with 1 or more	Mean	% with 1 or more	Mean
Untreated coronal caries	20.3	3.67	37.4	2.68	40.4	2.9	35.5	2.95

Results

Comparisons to Other Canadian Provinces

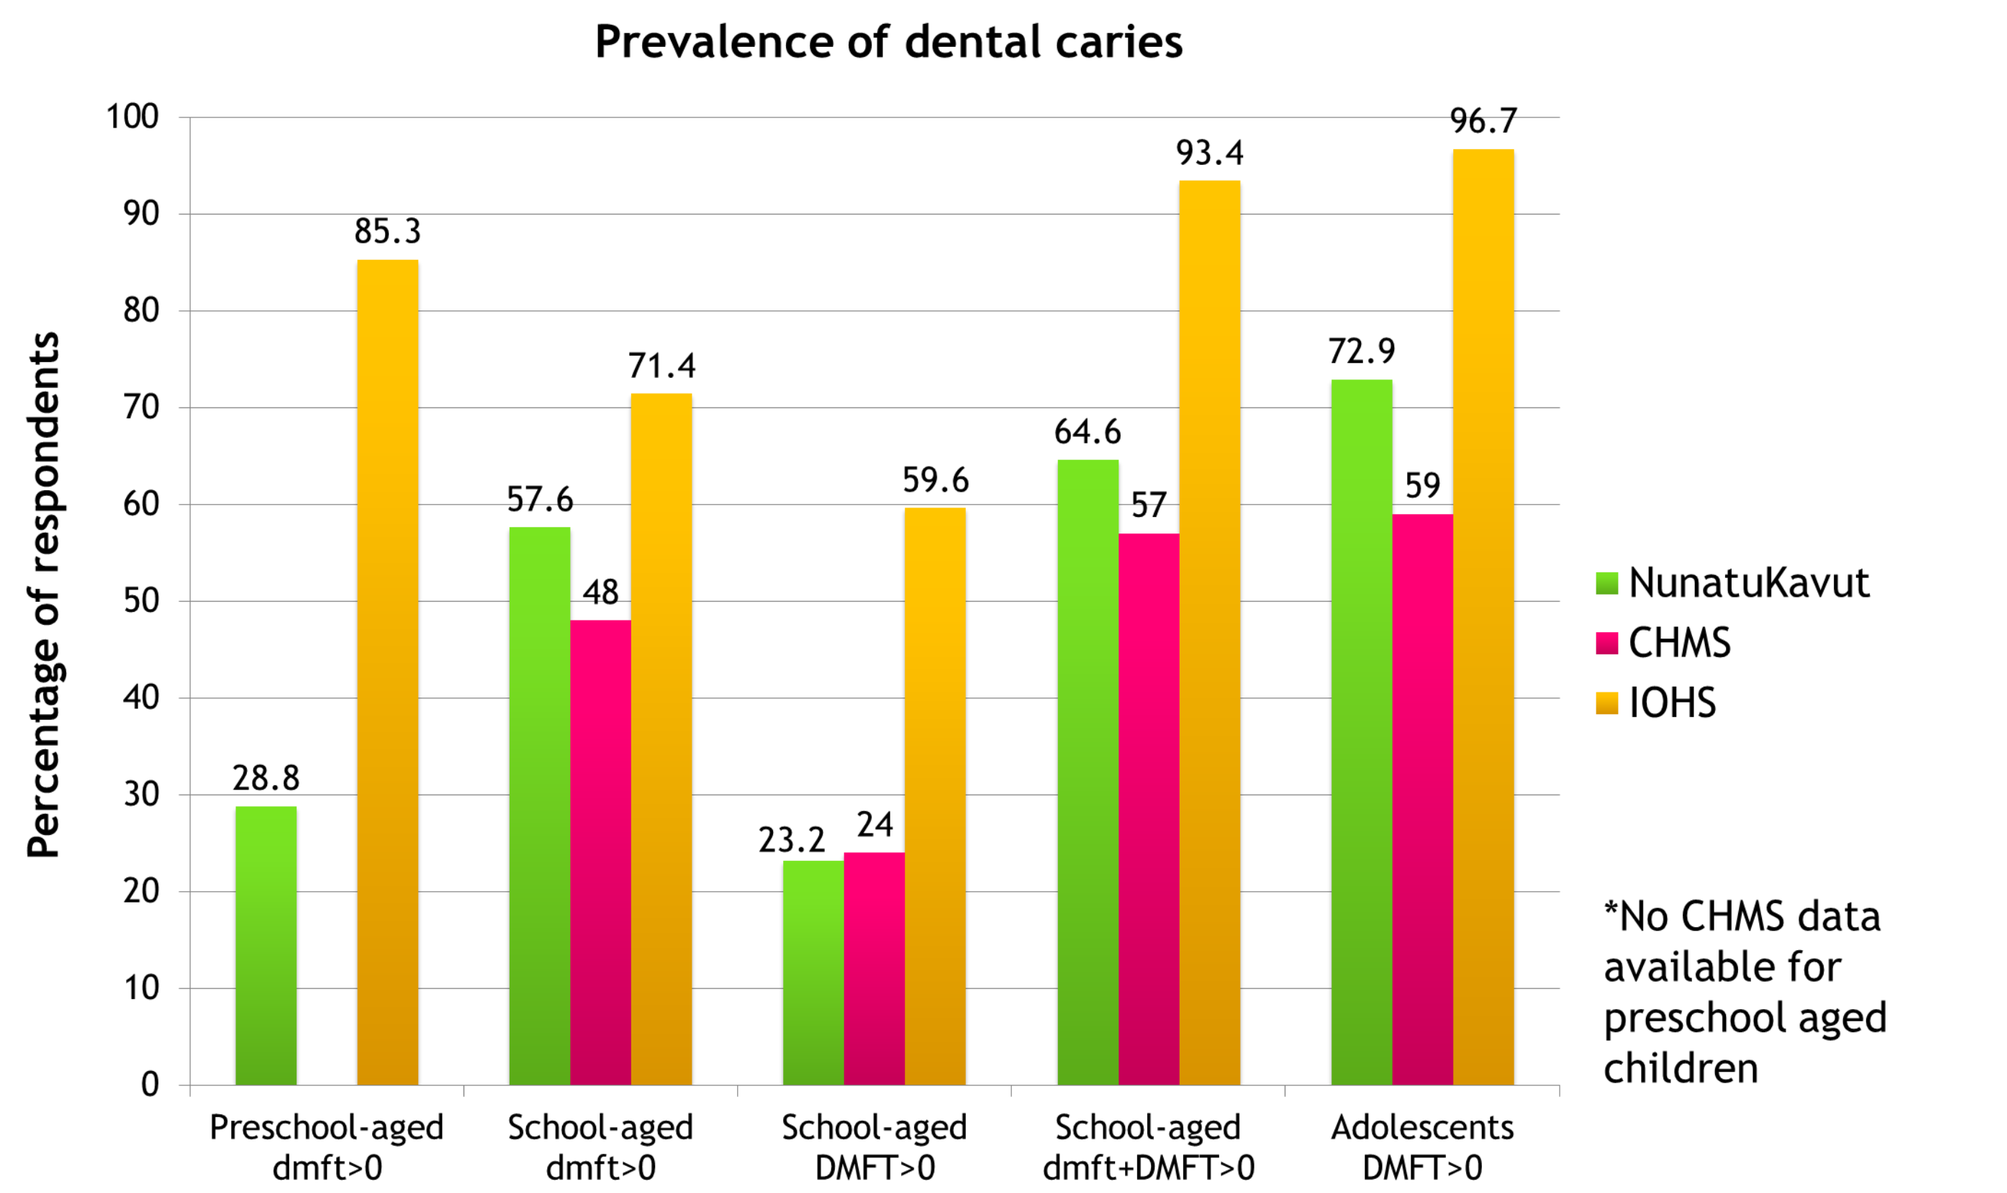
Prevalence of dental caries
Percentage of respondents
28.8
85.3
57.6
48
71.4
23.2
24
59.6
64.6
57
93.4
72.9
59
96.7
Preschool-aged dmft>0
School-aged dmft>0
School-aged DMFT>0
School-aged dmft+DMFT>0
Adolescents DMFT>0
NunatuKavut
CHMS
IOHS
*No CHMS data available for preschool aged children

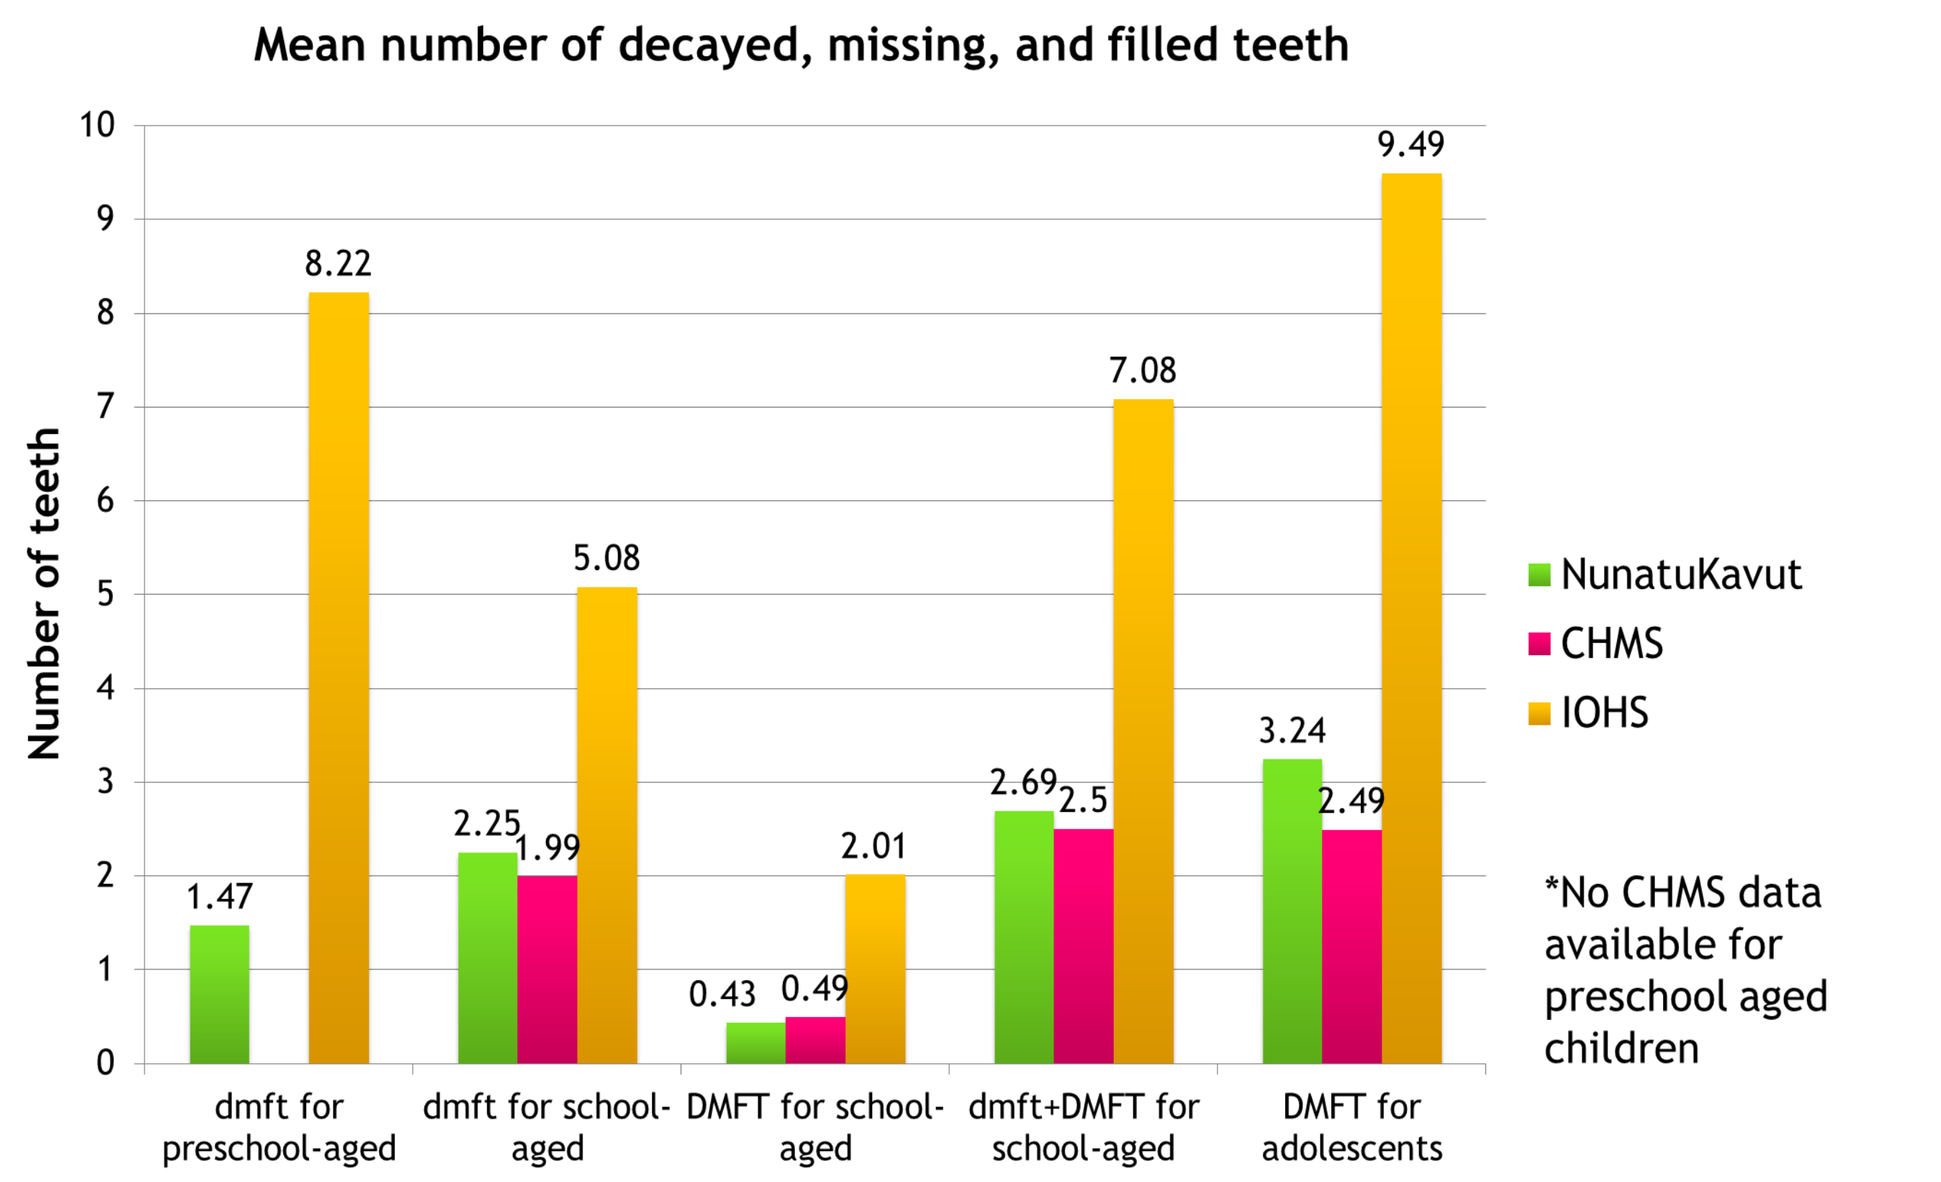
Mean number of decayed, missing, and filled teeth
Number of teeth
1.47
8.22
2.25
1.99
5.08
0.43
0.49
2.01
2.69
2.5
7.08
3.24
2.49
9.49
dmft for preschool-aged
dmft for school-aged
DMFT for school-aged
dmft+DMFT for school-aged
DMFT for adolescents
NunatuKavut
CHMS
IOHS
*No CHMS data available for preschool aged children

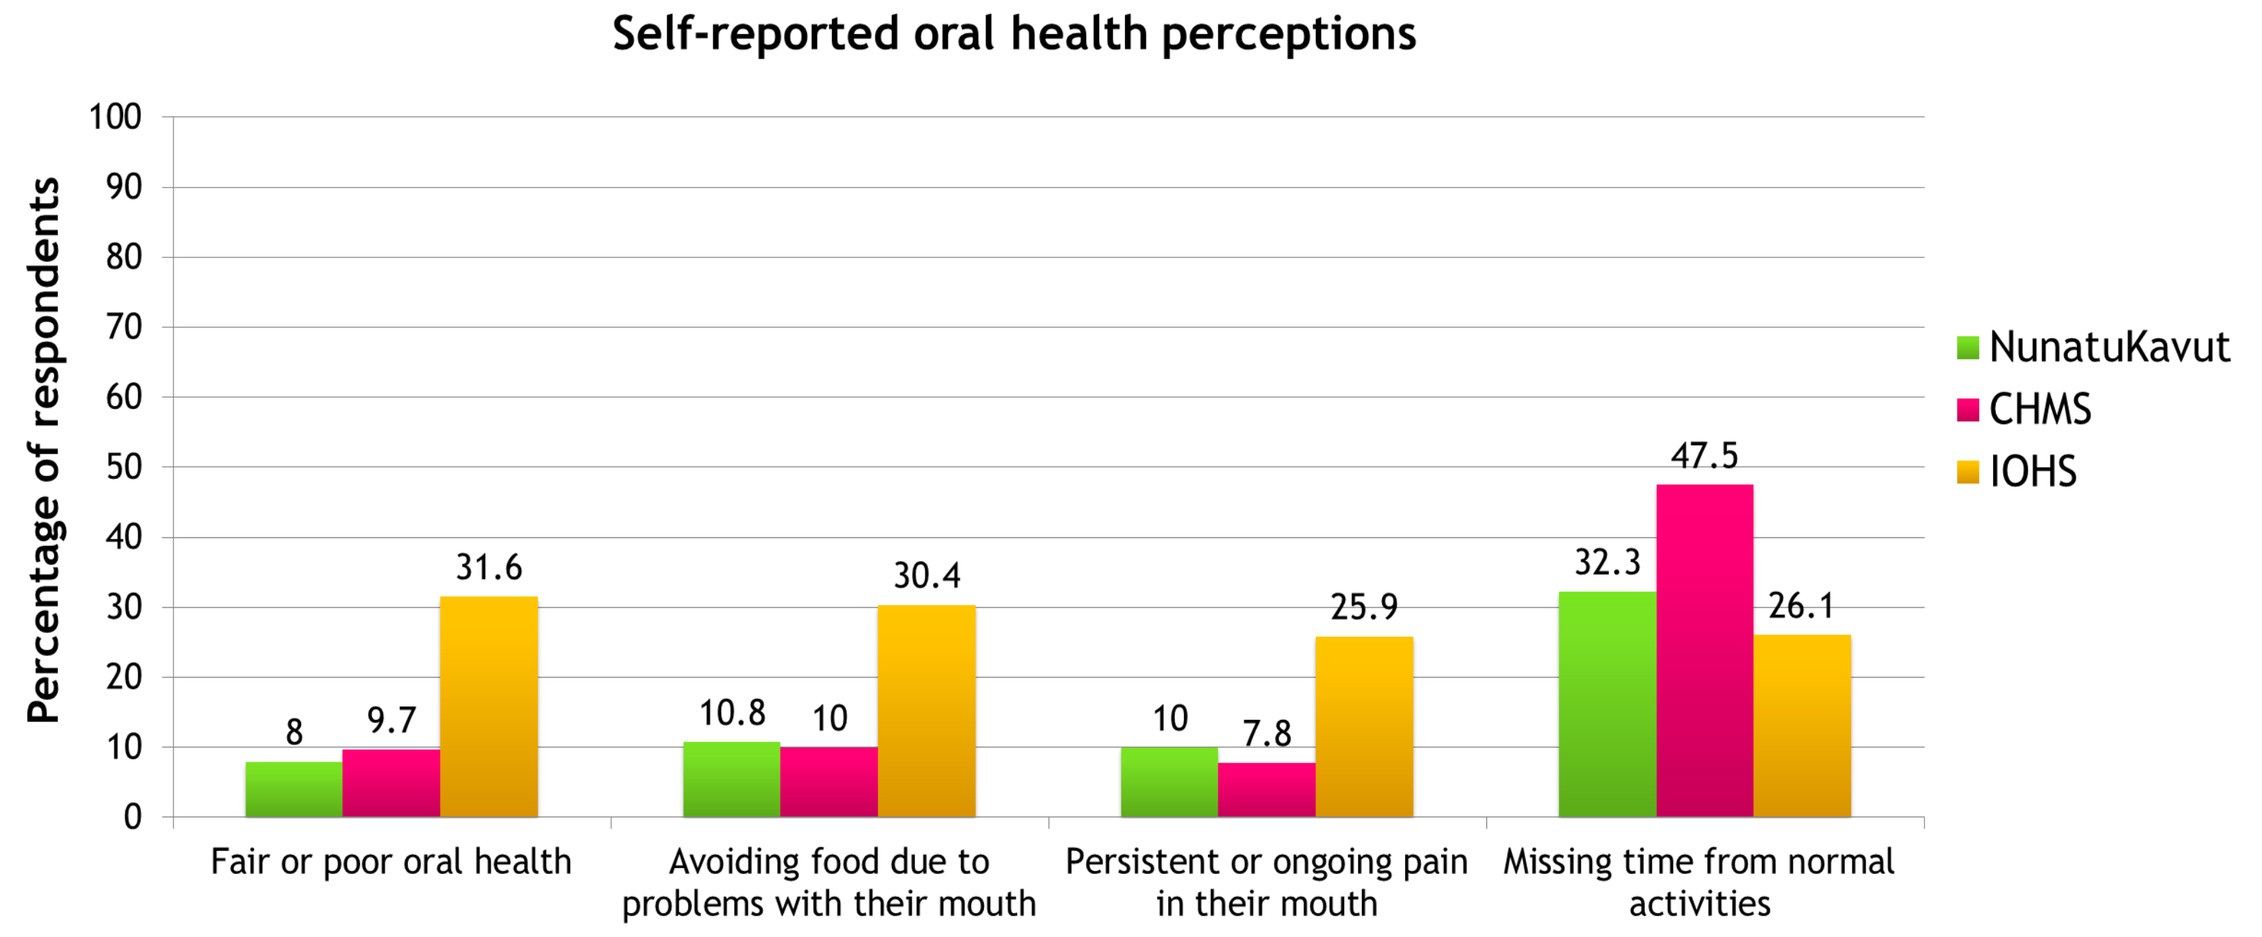
Self-reported oral health perceptions
Percentage of respondents
8
9.7
31.6
10.8
10
30.4
10
7.8
25.9
32.3
47.5
26.1
Fair or poor oral health
Avoiding food due to problems with their mouth
Persistent or ongoing pain in their mouth
Missing time from normal activities
NunatuKavut
CHMS
IOHS

Conclusions

1. Statistical comparisons to national data were not possible due to small population sizes. However, descriptive trends show that **compared to non-Aboriginal children and adolescents** in Canada, those in NunatuKavut:
 - *are equally satisfied with their oral health*
 - *have similar oral health care habits*
 - *have similar prevalence and severity of dental decay*
 - *are less likely to have visited a dental professional in the past year*
2. **Compared to other Inuit children and adolescents** in Canada, those in NunatuKavut:
 - *are more satisfied with their oral health*
 - *have better oral health care habits*
 - *have lower prevalence and severity of dental decay*
 - *have lower rates of untreated dental decay among adolescents*
3. The qualitative portion of this study explores possible underlying social, economic, historic and political explanations for these findings.